FACULTY OF BIOSCIENCE ENGINEERING

Effect of zinc oxide sources and dosages on intestinal Enterobacteriaceae and gut integrity of weaned piglets

J. Michiels[1*], N. Van Noten[1,2], J. Degroote[1,2], W. Wang[1,2] & A. Romeo[3]
[1] Department of Applied Biosciences, Faculty of Bioscience Engineering, Ghent University, Valentin Vaerwyckweg 1, 9000 Gent, Belgium; [2] Department of Animal Production, Faculty of Bioscience Engineering, Ghent University, Coupure Links 653, 9000 Gent, Belgium; [3] ANIMINE, 335 Chemin du Noyer, 74330 Sillingy, France

Conclusion: compared to regular ZnO at pharmacological dosage, the potentiated ZnO at low dosage showed equal reductions of Enterobacteriaceae counts, and improved gut epithelial barrier function and alkaline phosphatase mRNA expression in a similar or even better way

Background

Zn is an essential trace element for piglets
- Requirements NRC 2012; 100 and 80 mg Zn per kg feed for piglets of 5-11 and 11-25 kg BW, respectively
- Maximum levels are established to reduce output to environment and risk for co-selection for antibiotic resistance (e.g. in EU, 150 mg total Zn per kg feed for piglets)

ZnO is an antimicrobial growth promoter for weaned piglets
- Pharmacological doses: 2000-4000 mg Zn per kg feed
- Mode of action: improved performance, reduced incidence of diarrhoea, reduced inflammation, improved histo-morphology, improved tight junction integrity, reduced secretory responses, antimicrobial by free Zn^{2+} > radical inside the bacterium (*Lactobacillus* spp ↓, heterofermentative species ↑, clostridia ↑, diversity of *Enterobacteriaceae* ↑, *E. coli* in 1st week PW ↓), reduced adhesion of ETEC
- Drawbacks in practice: high buffering capacity, reduced feed intake, delayed *E. coli* infections, surge of *Streptococci* infections

Are there alternative ZnO sources at low dosage acting as growth promoter?
In this study, the effects of regular ZnO (110 and 2400 mg/kg of Zn) were compared to a potentiated ZnO source (HiZox®) at low dosage (110 and 220 mg/kg of Zn).

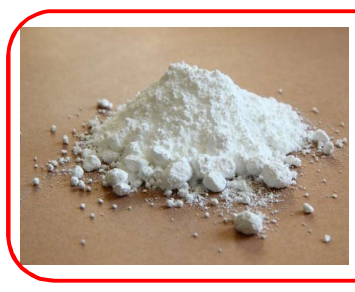

Regular ZnO

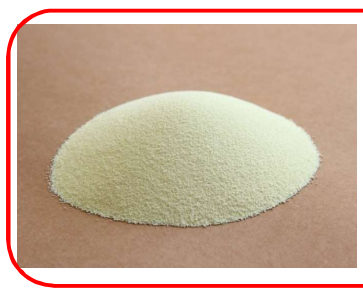

HiZox®, Animine
- Patented manufacturing technology
- Unique physico-chemical features, including high purity standards, high specific surface area due to high porosity, optimal particle size distribution

Materials and Methods

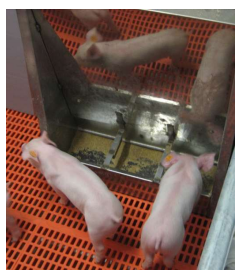

Animals and diets
- 48 weaned piglets, 19 d, 6.04±0.77 kg
- 24 pens with 2 piglets each (balanced for litter and gender)
- 6 treatments replicated in 4 pens
- Basal diet was free of supplemental Zn, low Cu, no organic acids
- High iron level was used to induce gastro-intestinal disturbances
- Animals were fed for 15 days, and then euthanised and sampled for gut health parameters

Dietary treatments

	T1	T2	T3	T4	T5	T6
Fe from $FeSO_4$ (mg/kg)	100	100	500	500	500	500
Zn from ZnO (mg/kg)	110	2400	110	2400	110	220
Source of ZnO	regular ZnO				HiZox	

Measurements
- Animal performance (n=4) and health (including fecal scores) (n=8)
- Digesta from stomach, proximal (first 25% of length) and distal (last 25% of length) small intestine (n=8)
 - ✓ pH
 - ✓ Dry matter
 - ✓ *E. coli* and coliform counts by plating on selective media
 - ✓ Lactobacilli, *Clostridrium* XIVa, Enterobacteriaceae by qPCR (only T3-T6)
 - ✓ Diversity indices (Richness and Shannon) (only T3-T6)
- Small intestinal mucosa at 75% of length (n=8)
 - ✓ *Ex vivo* paracellular with FITC-4kDa as marker
 - ✓ Transepithelial electrical resistance (TEER)
 - ✓ Basal short-circuit current (Isc) and Isc upon stimulation with the secretagogues serotonin and theophylline
 - ✓ Gene expression for (only T3-T6):
 - o Tight junctions proteins: occludin, claudin 1, claudin 5, claudin 7 and zona occludens
 - o Toll like receptor 4
 - o Inflammatory cytokines: TNFα, IFNγ and Il1β
 - o Intestinal alkaline phosphatase

Statistics
Mixed model with factor treatment as fixed factor and factor block as random factor and their interaction (SAS Entreprise Guide 8.0). Means are separated by the Tukey-test (P<0.05)

Results

- In contrast to anticipated, the high Fe level did not deteriorate performances; here below only results of T3-T6 are then given
- HiZox at 220 mg Zn/kg improved performance in a almost similar way as pharmacological dose of regular ZnO
- Non-significant increase of gastric pH with pharmalogical dose of regular ZnO
- Bacteria by plating: regular ZnO at pharmacological dose and HiZox at both 110 and 220 mg Zn/kg reduced numbers of *E. coli* and coliforms by ≥ 1 log_{10} CFU/g in distal small intestine (Fig. 1); smaller and non-significant reductions in proximal small intestine
- Bacteria by qPCR: data suggest no effect on Lactobacilli, reduced *Clostridium* XIVa by HiZox at 110 mg Zn/kg and reduced Enterobacteriaceae by conventional ZnO at pharmacological dose and HiZox

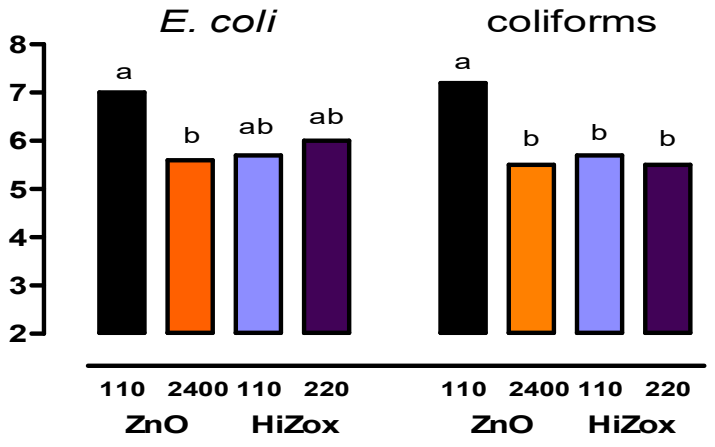
E. coli
coliforms
8
7
6
5
4
3
2
a
b
ab
ab
a
b
b
b
110 2400 110 220
ZnO HiZox
110 2400 110 220
ZnO HiZox

Fig 1. Effect of zinc sources and dosages on distal small intestinal E. coli and coliforms numbers (log_{10} CFU/g)

- HiZox improved significantly barrier function shown by increased TEER (Fig 2.)
- Tendencies indicate increased mRNA expression of tight junction proteins claudin 1 and zona occludens 1 in pharmacological dose of regular ZnO and HiZox, consistent with increased TEER values
- HiZox at 220 mg Zn/kg increased mRNA level of intestinal alkaline phosphatase, an important defence factor for mucosal homeostasis (Fig 3.)

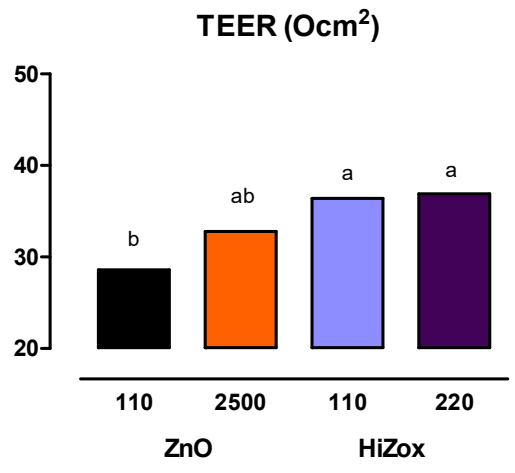
TEER (Ocm2)
50
40
30
20
b
ab
a
a
110
2500
110
220
ZnO
HiZox

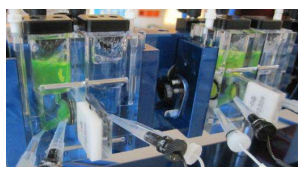

Fig 2. Effect of zinc sources and dosages on distal small intestinal mucosal barrier function by measuring transepithelial resistance (TEER)

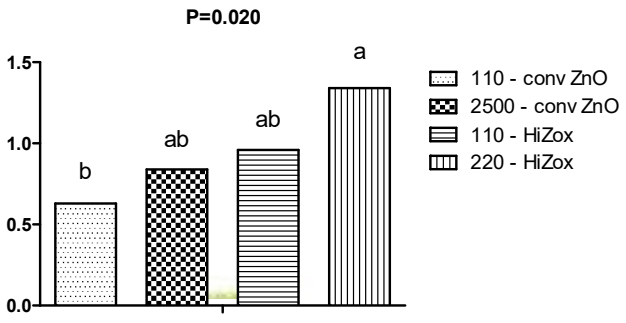
P=0.020
1.5
1.0
0.5
0.0
b
ab
ab
a
110 - conv ZnO
2500 - conv ZnO
110 - HiZox
220 - HiZox

Fig 3. Effect of zinc sources and dosages on distal small intestinal mucosal mRNA level of intestinal alkaline phosphatase